Evaluation of AB-101, an Allogeneic Cord Blood-derived Natural Killer (NK) Cell Therapy, as an ADCC Enhancer in Hematologic and Solid Tumors

Paul Rogers[1], Bitna Yang[2], Hyojin Kim[2], Yusun Kim[2], Sunglim Cho[2], Hyeong Jin Nam[2], Bret Morin[1], Amanda Conerty[1],Kumar Karyampudi[1], Lisa Guerrettaz[1] , Peter Flynn[1], Bokyung Min[2] and Heather Raymon[1]
[1]Artiva Biotherapeutics, San Diego, CA [2]GC Cell, Yongin-si, South Korea

Introduction

AB-101 is a non-engineered, allogeneic, off-the-shelf, cryopreserved cord blood-derived natural killer (NK)-cell therapy in development as a cancer therapeutic. A highly scaled manufacturing process enables production of 1000s of doses from a single donor cord blood unit (CBU). AB-101 has been optimized for combination with monoclonal antibodies (mAbs) to enhance antibody-dependent cellular cytotoxicity (ADCC) and anti-tumor responses through selection of key attributes in the CBU. These include a KIR-B haplotype[1] and natural high-affinity variant of CD16 (158V/V polymorphism)[2], which are associated with improved anti-tumor activity and ADCC enhancement. Administration of AB-101 to patients in combination with mAbs has the potential to enhance the ADCC response, thereby increasing anti-tumor activity. Here we present preclinical data to support development of AB-101 in combination with mAbs as an ADCC enhancer in hematologic malignancies and solid tumor indications.

Methods

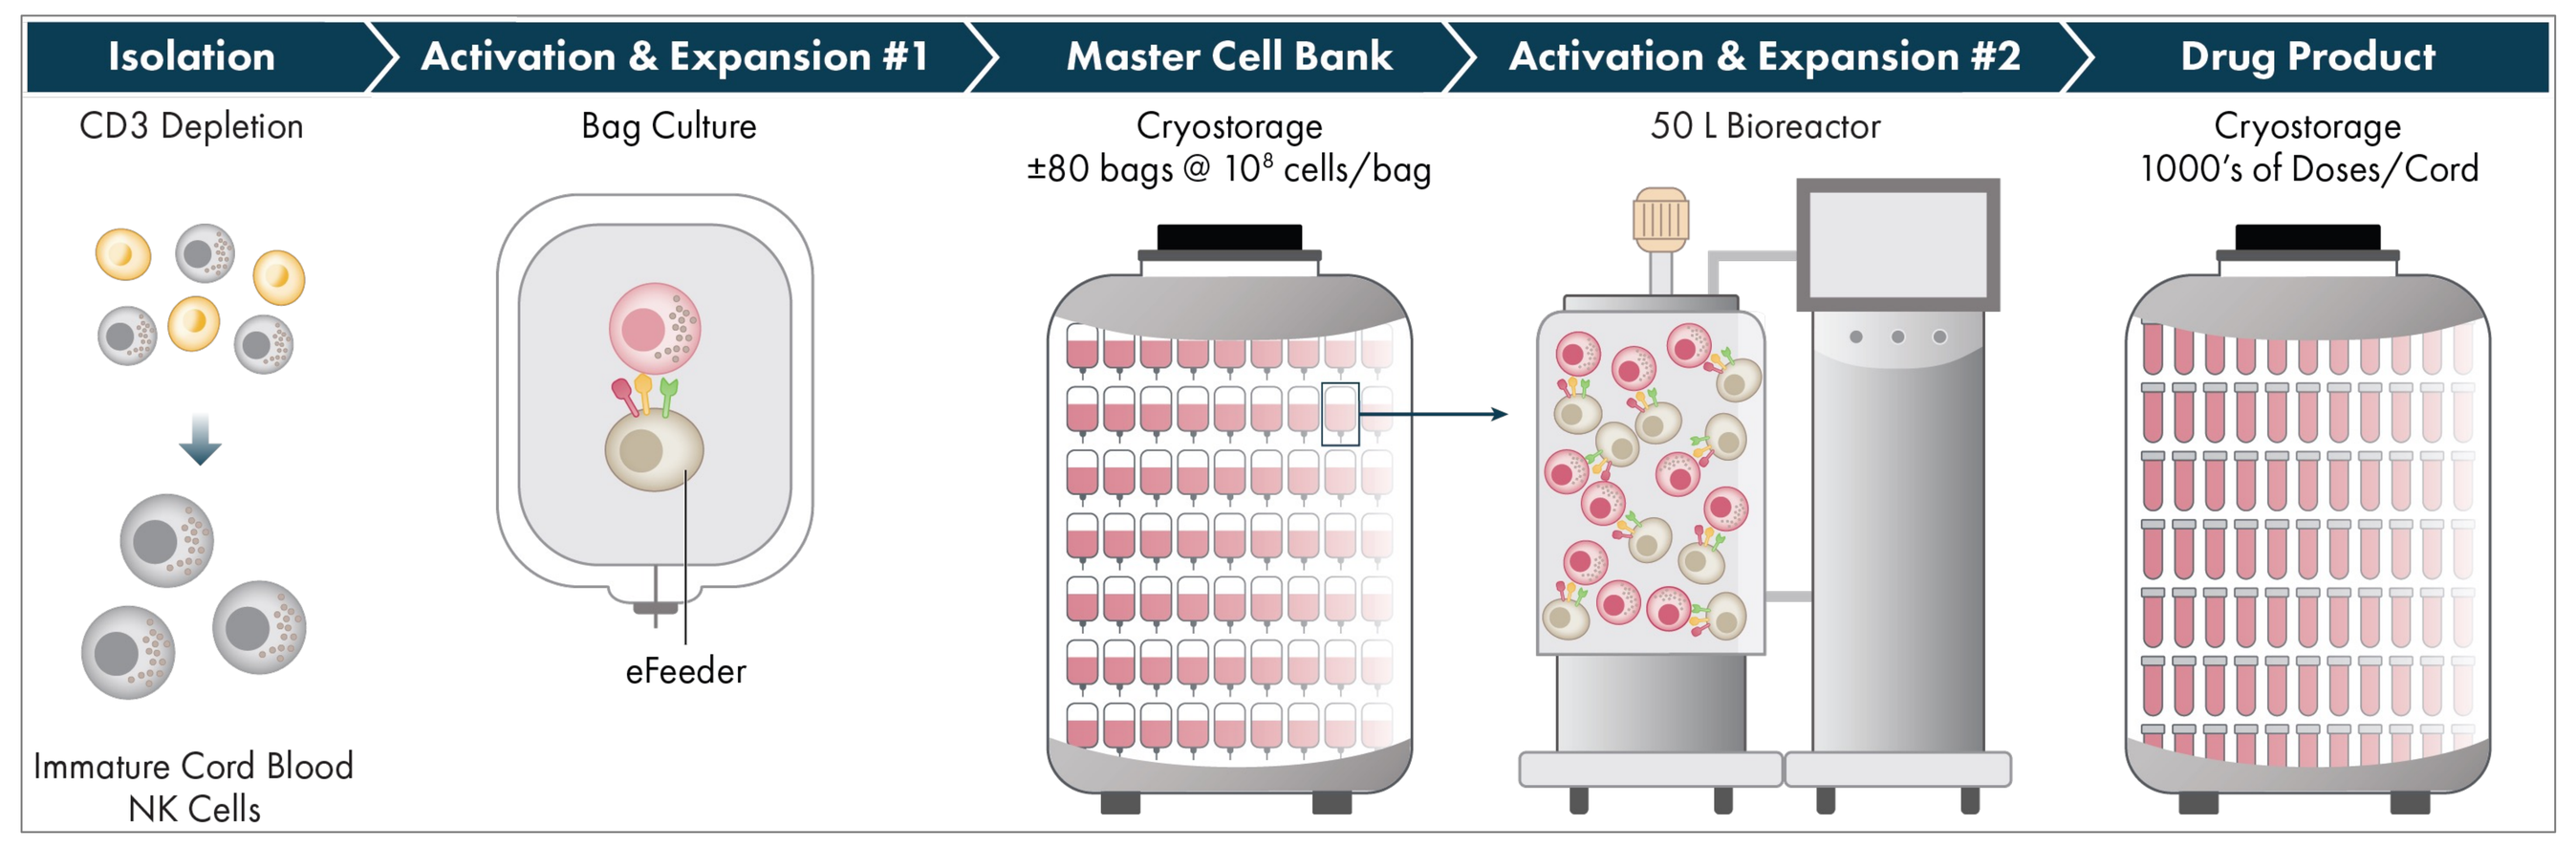
Isolation
Activation & Expansion #1
Master Cell Bank
Activation & Expansion #2
Drug Product
CD3 Depletion
Bag Culture
Cryostorage
±80 bags @ 10⁸ cells/bag
50 L Bioreactor
Cryostorage
1000's of Doses/Cord
eFeeder
Immature Cord Blood NK Cells

AB-101 manufacturing- Uses U.S. licensed umbilical cord blood (CB) units with selected characteristics. Following isolation, NK cells are expanded utilizing a proprietary engineered feeder cell (eFeeder) used to derive a master cell bank, followed by a bioreactor-based large-scale NK cell expansion and activation process to produce pure NK cells. High and consistent expression of the 158V/V CD16 is achieved in the AB-101 drug product, without the requirement for engineering of the cells. The scale of production potentially enables hundreds to thousands of patients to be treated from a single donor CBU.

In vitro characterization of AB-101 included evaluation of the purity and expression of cell surface markers by flow cytometry. In vitro ADCC assays were performed at various effector to target ratios against hematologic (Raji, ARH-77, Ramos) tumors in combination with Rituximab or Obinutuzumab. In addition, AB-101 efficacy was assessed in vivo in B-cell lymphoma (Raji, Ramos) xenograft models. These data supported the initiation of a clinical trial (NCT04673617) assessing AB-101 as monotherapy and in combination with Rituximab in patients with Relapsed/Refractory B-Cell Non-Hodgkin Lymphoma.

AB-101 was also characterized in solid tumor cell lines (Breast-MDA-MB-468, Ovarian-SKOV-3) in the presence of approved therapeutic antibodies (Cetuximab and Trastuzumab) demonstrating the potential for additional combinations.

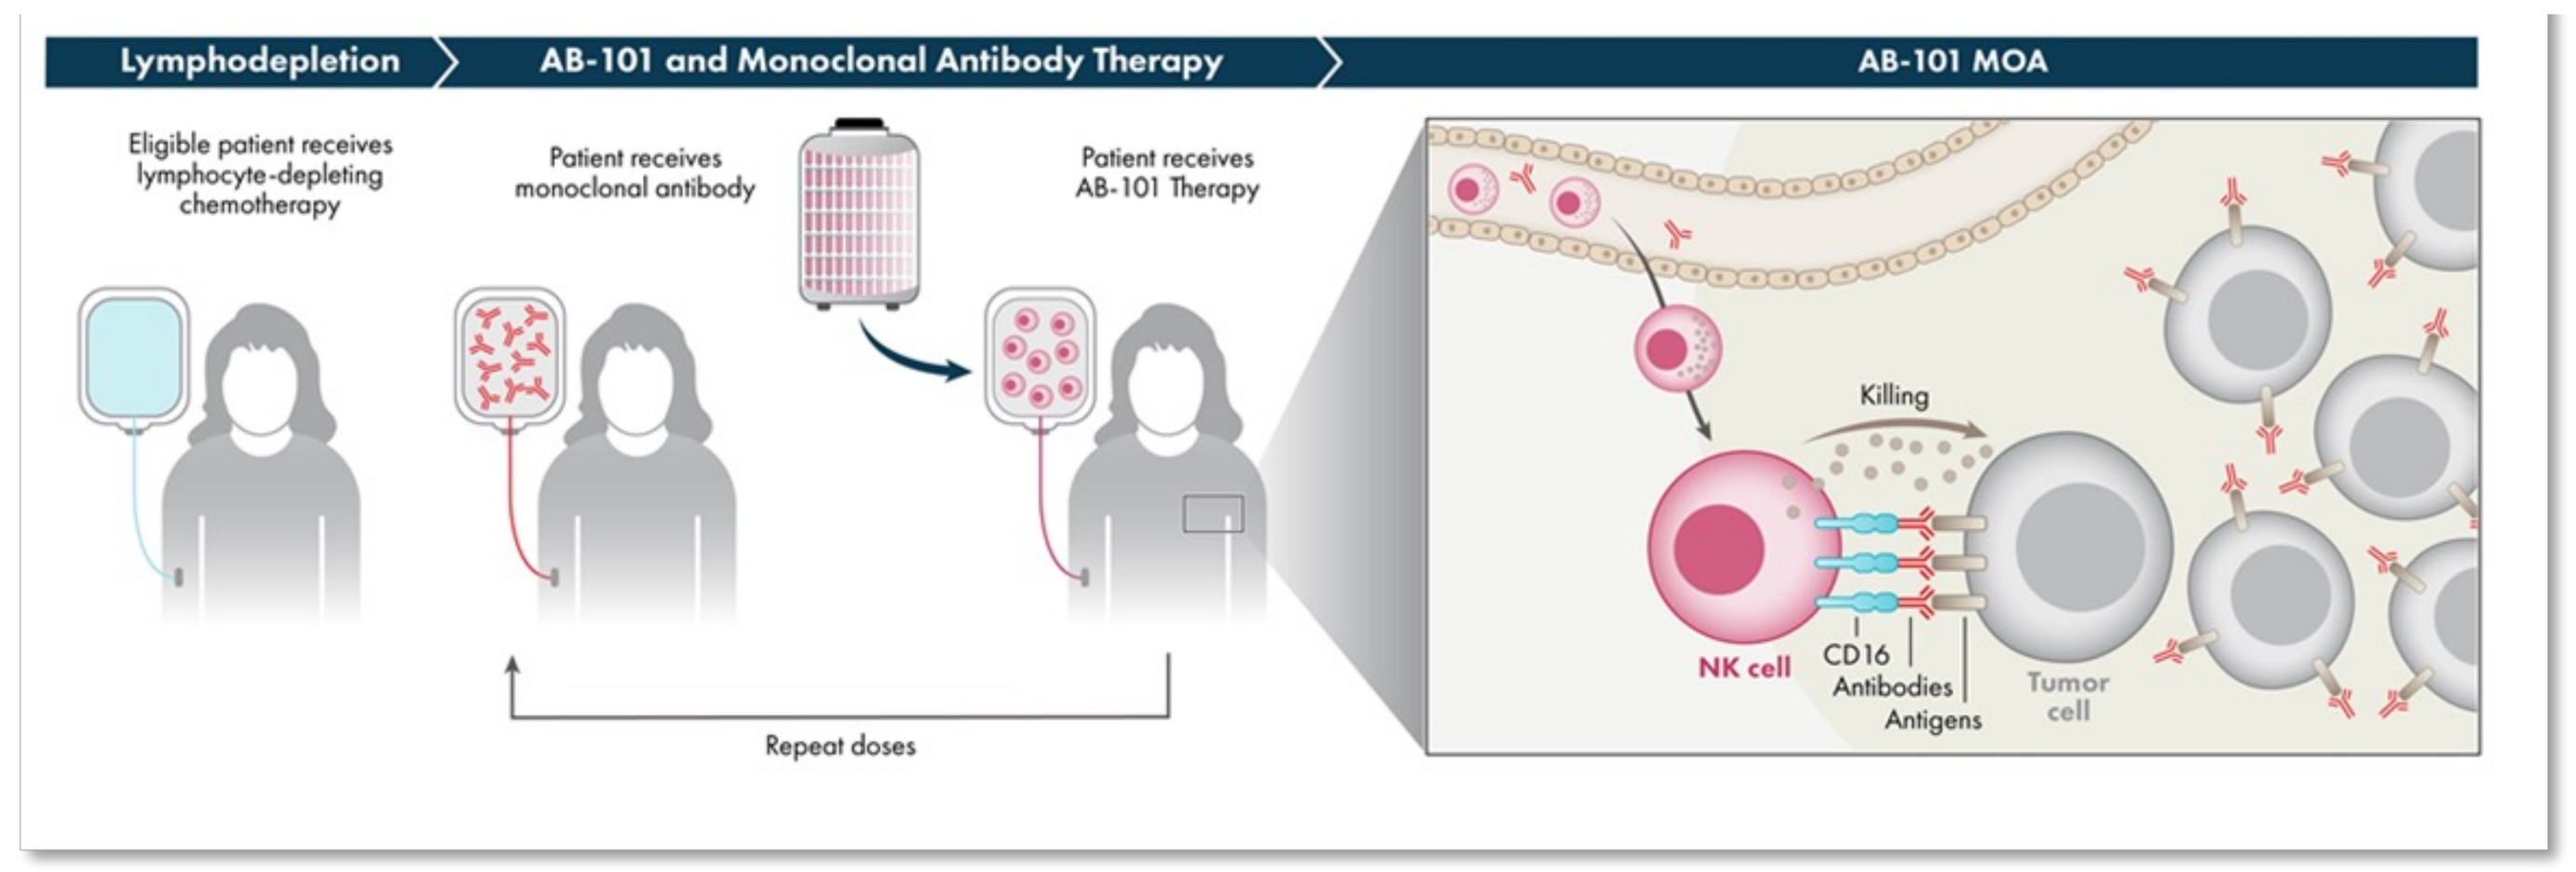
Lymphodepletion
AB-101 and Monoclonal Antibody Therapy
AB-101 MOA
Eligible patient receives lymphocyte-depleting chemotherapy
Patient receives monoclonal antibody
Patient receives AB-101 Therapy
Repeat doses
Killing
NK cell
CD16
Antibodies
Antigens
Tumor cell

AB-101 Mechanism of Action as an ADCC Enhancer – NK cells that can enhance a patient's ADCC, or antibody-dependent cellular cytotoxicity, response when undergoing monoclonal antibody therapy for either hematological or solid tumors.

Results

Figure 1. Cord blood has significant advantage over peripheral blood as NK source

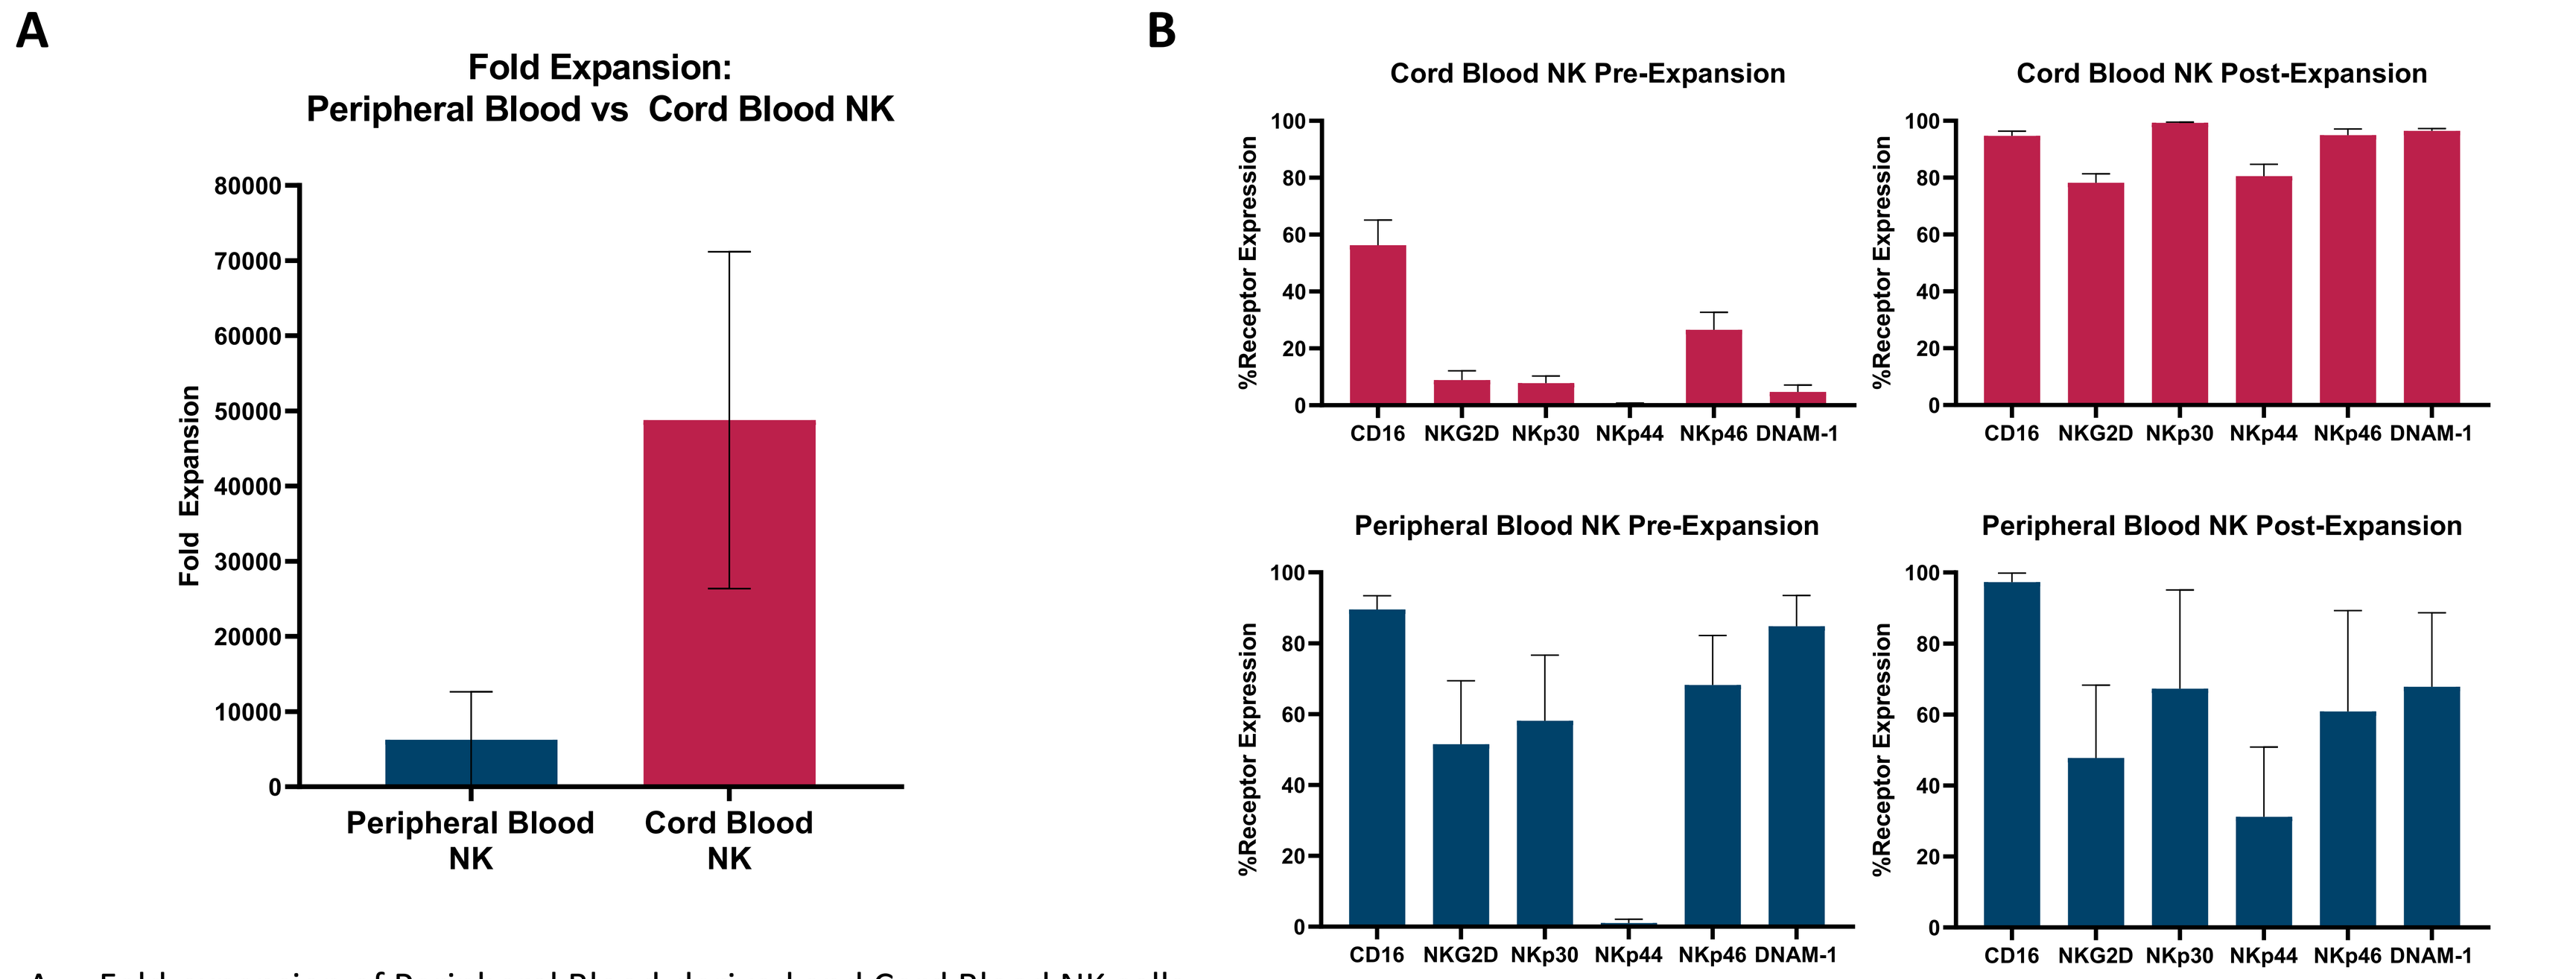
A
Fold Expansion: Peripheral Blood vs Cord Blood NK
Fold Expansion
Peripheral Blood NK
Cord Blood NK
B
Cord Blood NK Pre-Expansion
Cord Blood NK Post-Expansion
Peripheral Blood NK Pre-Expansion
Peripheral Blood NK Post-Expansion
%Receptor Expression
CD16
NKG2D
NKp30
NKp44
NKp46
DNAM-1

A. Fold-expansion of Peripheral Blood-derived and Cord Blood NK cells.
B. Frequency of cell surface expression of NK receptors on Cord Blood NK cells pre- (n=3) and post-expansion (n=8) compared to Peripheral Blood NK cells pre- (n= 17) and post- (n=40) expansion. Data are plotted as the mean ± SD.

Figure 2. AB-101: purity and phenotypic characterization

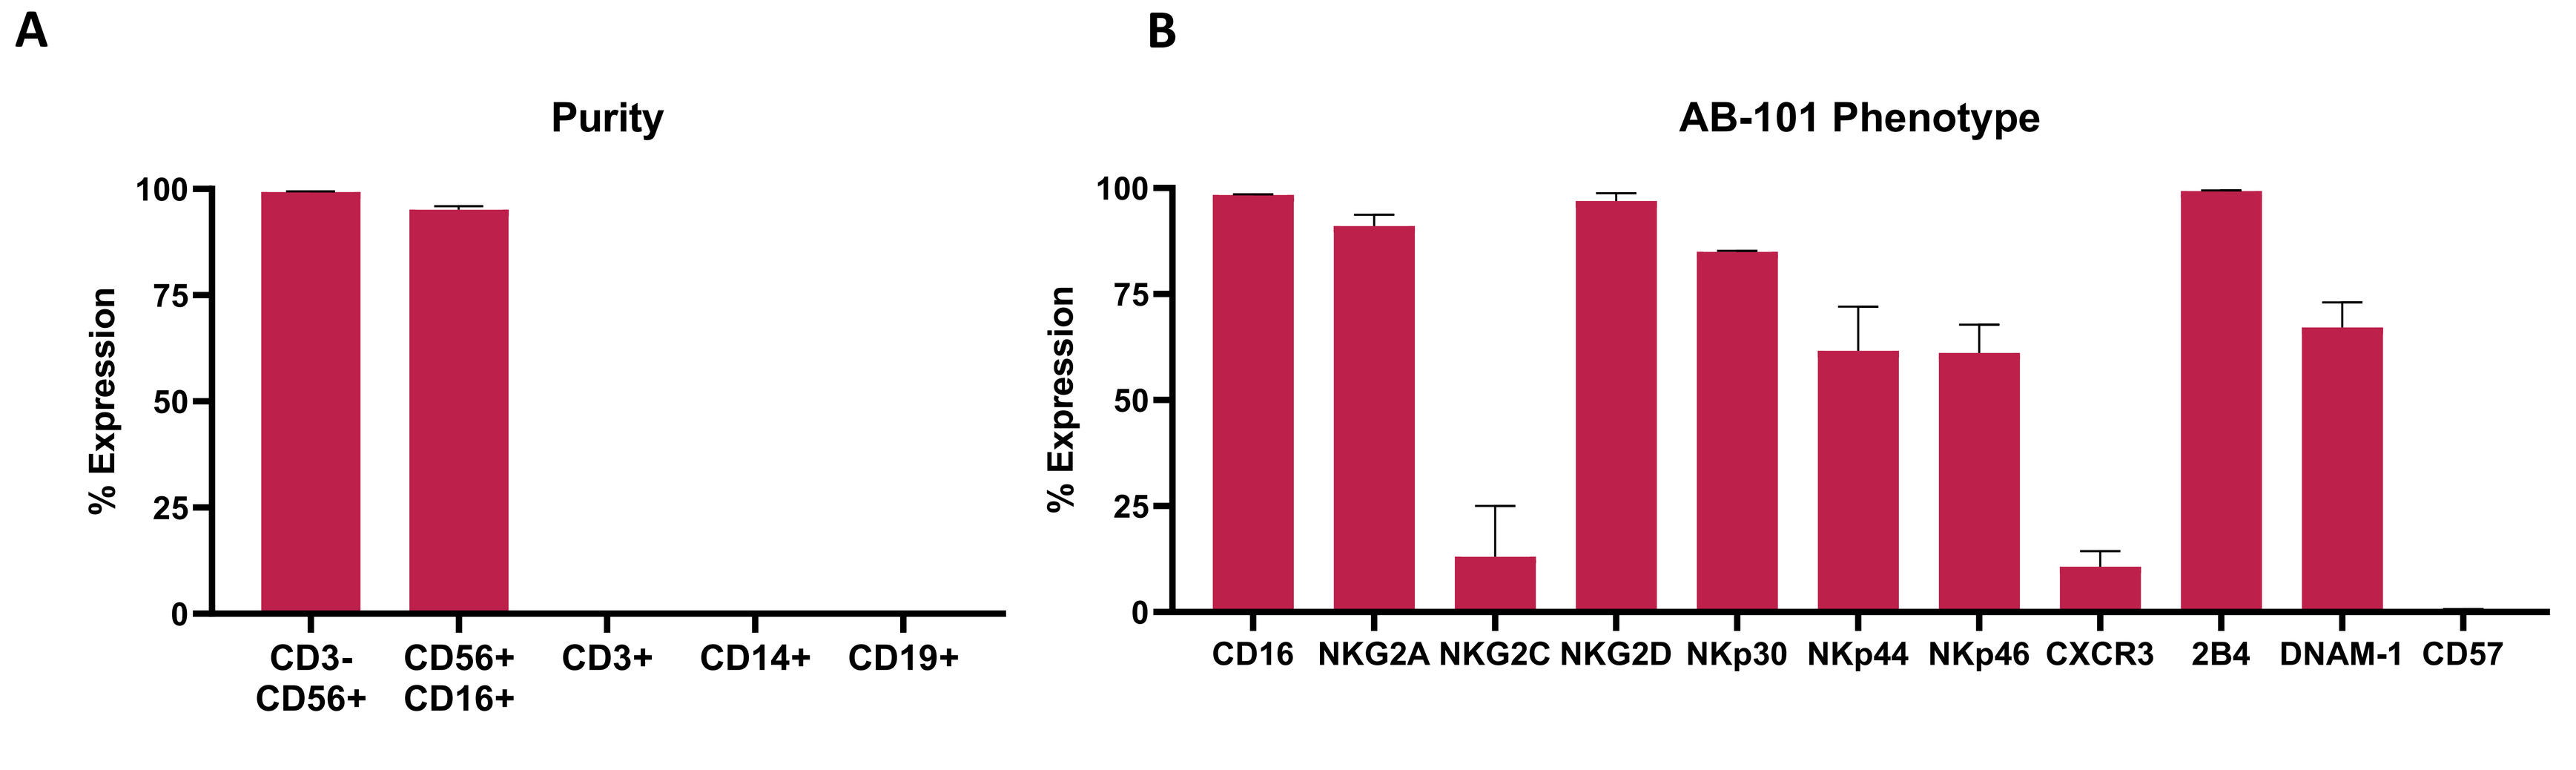
A
Purity
% Expression
CD3-CD56+
CD56+ CD16+
CD3+
CD14+
CD19+
B
AB-101 Phenotype
% Expression
CD16
NKG2A
NKG2C
NKG2D
NKp30
NKp44
NKp46
CXCR3
2B4
DNAM-1
CD57

A. AB-101 characterization and frequency of NK cells (CD3-CD56+), (CD16+CD56+).
B. Frequency of cell surface expression of NK receptors was assessed on AB-101 gating on CD3-CD56+ cells. Data are plotted as the mean ± SEM.

Figure 3. Enhanced ADCC with AB-101 + anti-CD20 mAbs, Rituximab and Obinutuzumab

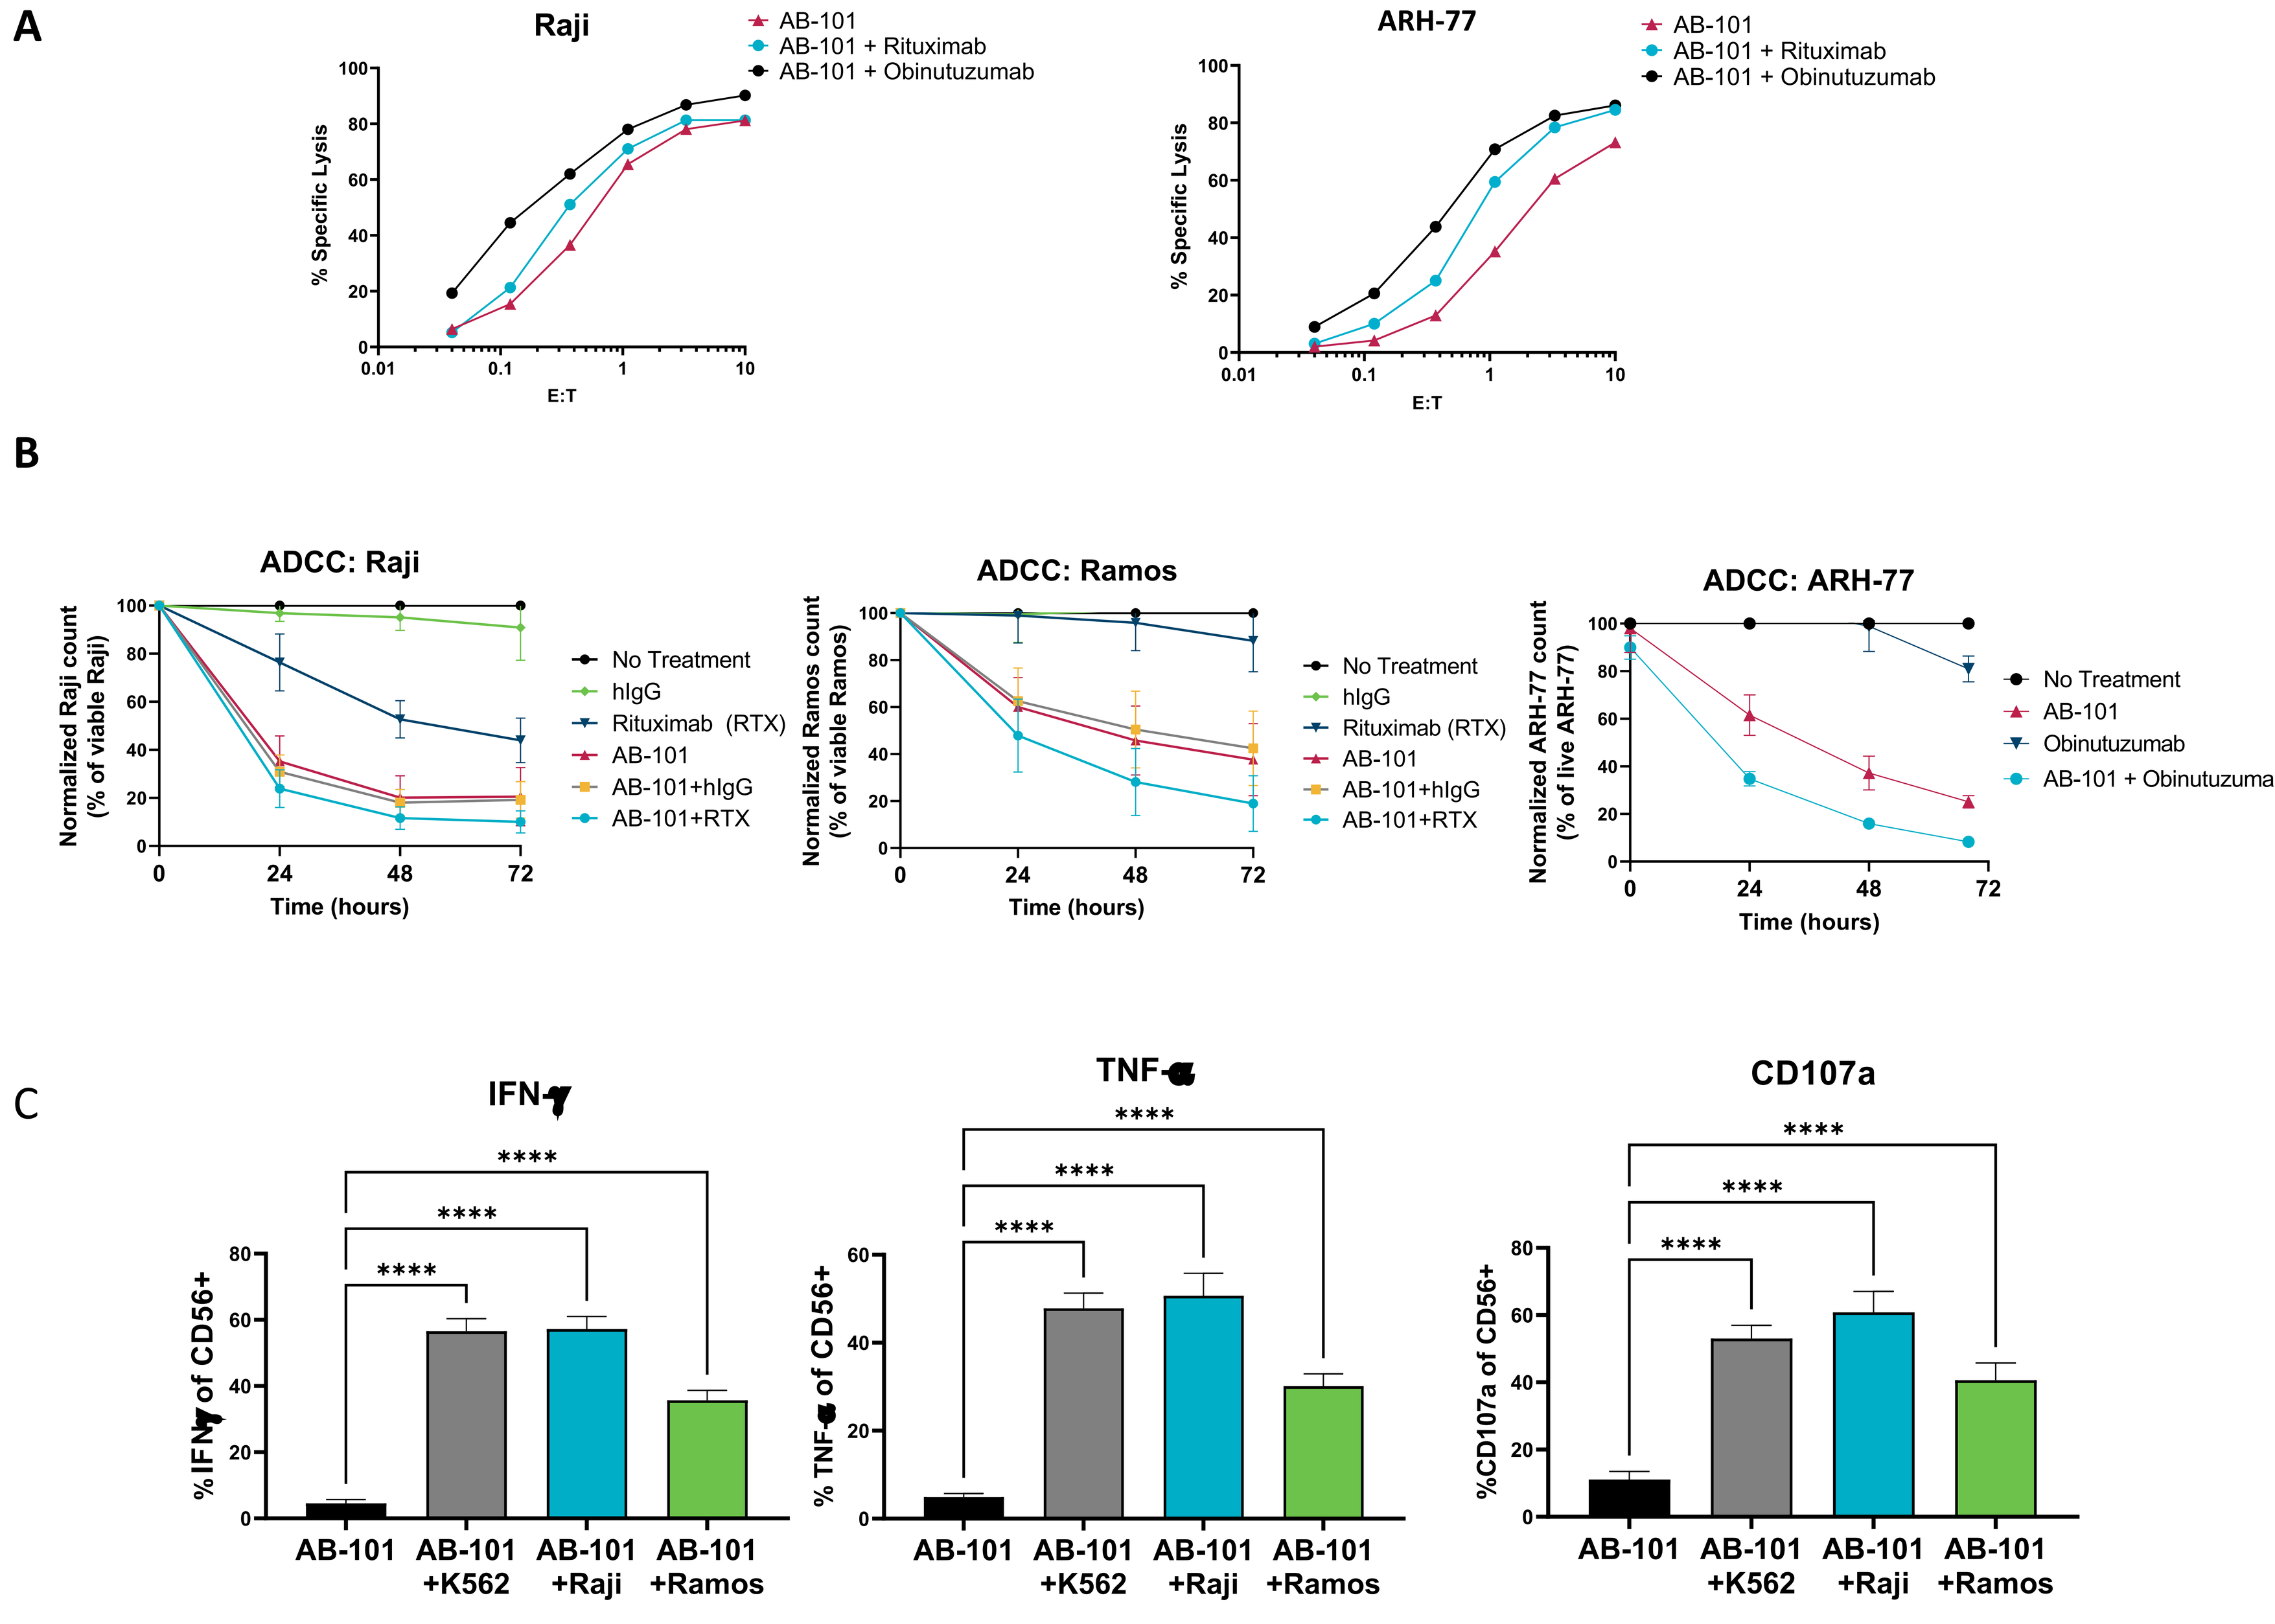
A
Raji
ARH-77
AB-101
AB-101 + Rituximab
AB-101 + Obinutuzumab
% Specific Lysis
E:T
B
ADCC: Raji
ADCC: Ramos
ADCC: ARH-77
No Treatment
hIgG
Rituximab (RTX)
AB-101
AB-101+hIgG
AB-101+RTX
Obinutuzumab
AB-101 + Obinutuzumab
Normalized Raji count (% of viable Raji)
Normalized Ramos count (% of viable Ramos)
Normalized ARH-77 count (% of live ARH-77)
Time (hours)
C
CD107a
%CD107a of CD56+

AB-101
AB-101 +K562
AB-101 +Raji
AB-101 +Ramos

A. Cytotoxicity of AB-101 alone and in combination with anti-CD20 monoclonal antibodies (Rituximab and Obinutuzumab) against lymphoma cell lines.
B. Long-term cellular cytotoxicity of AB-101 alone and in combination with Rituximab or Obinutuzumab or isotype control (hIgG) against CD20+ tumor cell lines (Raji, Ramos and ARH-77) over 72hours at a 1:1 Effector:Target (E:T) ratio.
C. AB-101 was stimulated for 4 hours with tumor targets at a 1:1 E:T ratio. Following stimulation, cells were collected, and degranulation (CD107a) and cytokine secretion were analyzed by flow cytometry. Plots show the mean ± SEM.

Figure 4. AB-101 demonstrates enhanced in vivo anti-tumor activity

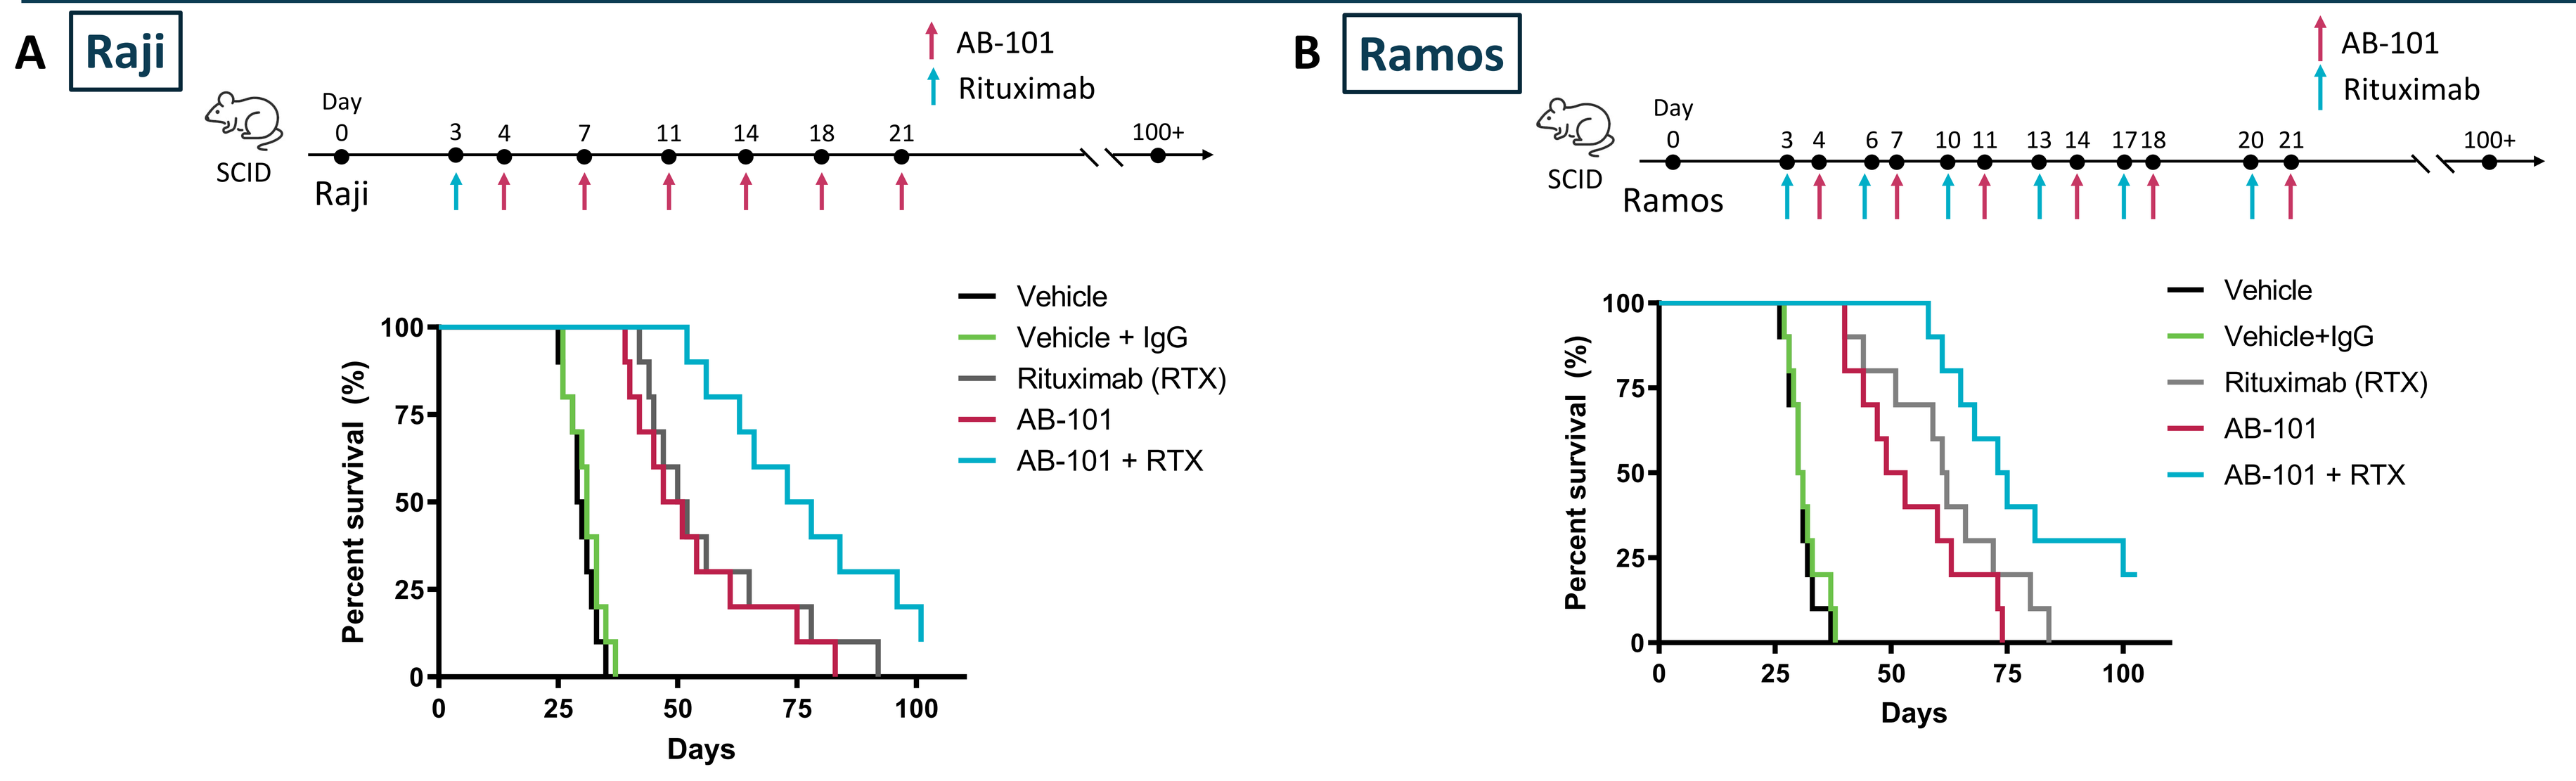
A
Raji
B
Ramos
AB-101
Rituximab
SCID
Day
100+
Vehicle
Vehicle + IgG
Rituximab (RTX)
AB-101
AB-101 + RTX
Percent survival (%)
Days

A. Experimental design: Raji B-lymphoma model (IV) in SCID mice administered vehicle, vehicle + IgG (0.01µg), Rituximab (0.01µg), AB-101 ($2x10^7$), or AB-101 ($2x10^7$) + Rituximab (0.01µg). A total of 6 doses of AB-101 and 1 dose of Rituximab were given. Kaplan-Meier survival curve is representative of % survival in each group.
B. Experimental design: Ramos lymphoma model (IV) in SCID mice administered vehicle, vehicle + IgG (0.01µg), Rituximab (0.3µg), AB-101 ($2x10^7$), or AB-101 ($2x10^7$) + Rituximab (0.3µg). A total of 6 doses of AB-101 and 6 doses of Rituximab were given. Kaplan-Meier survival curve is representative of % survival rate in each group.

Figure 5. AB-101 demonstrates enhanced ADCC in solid tumors

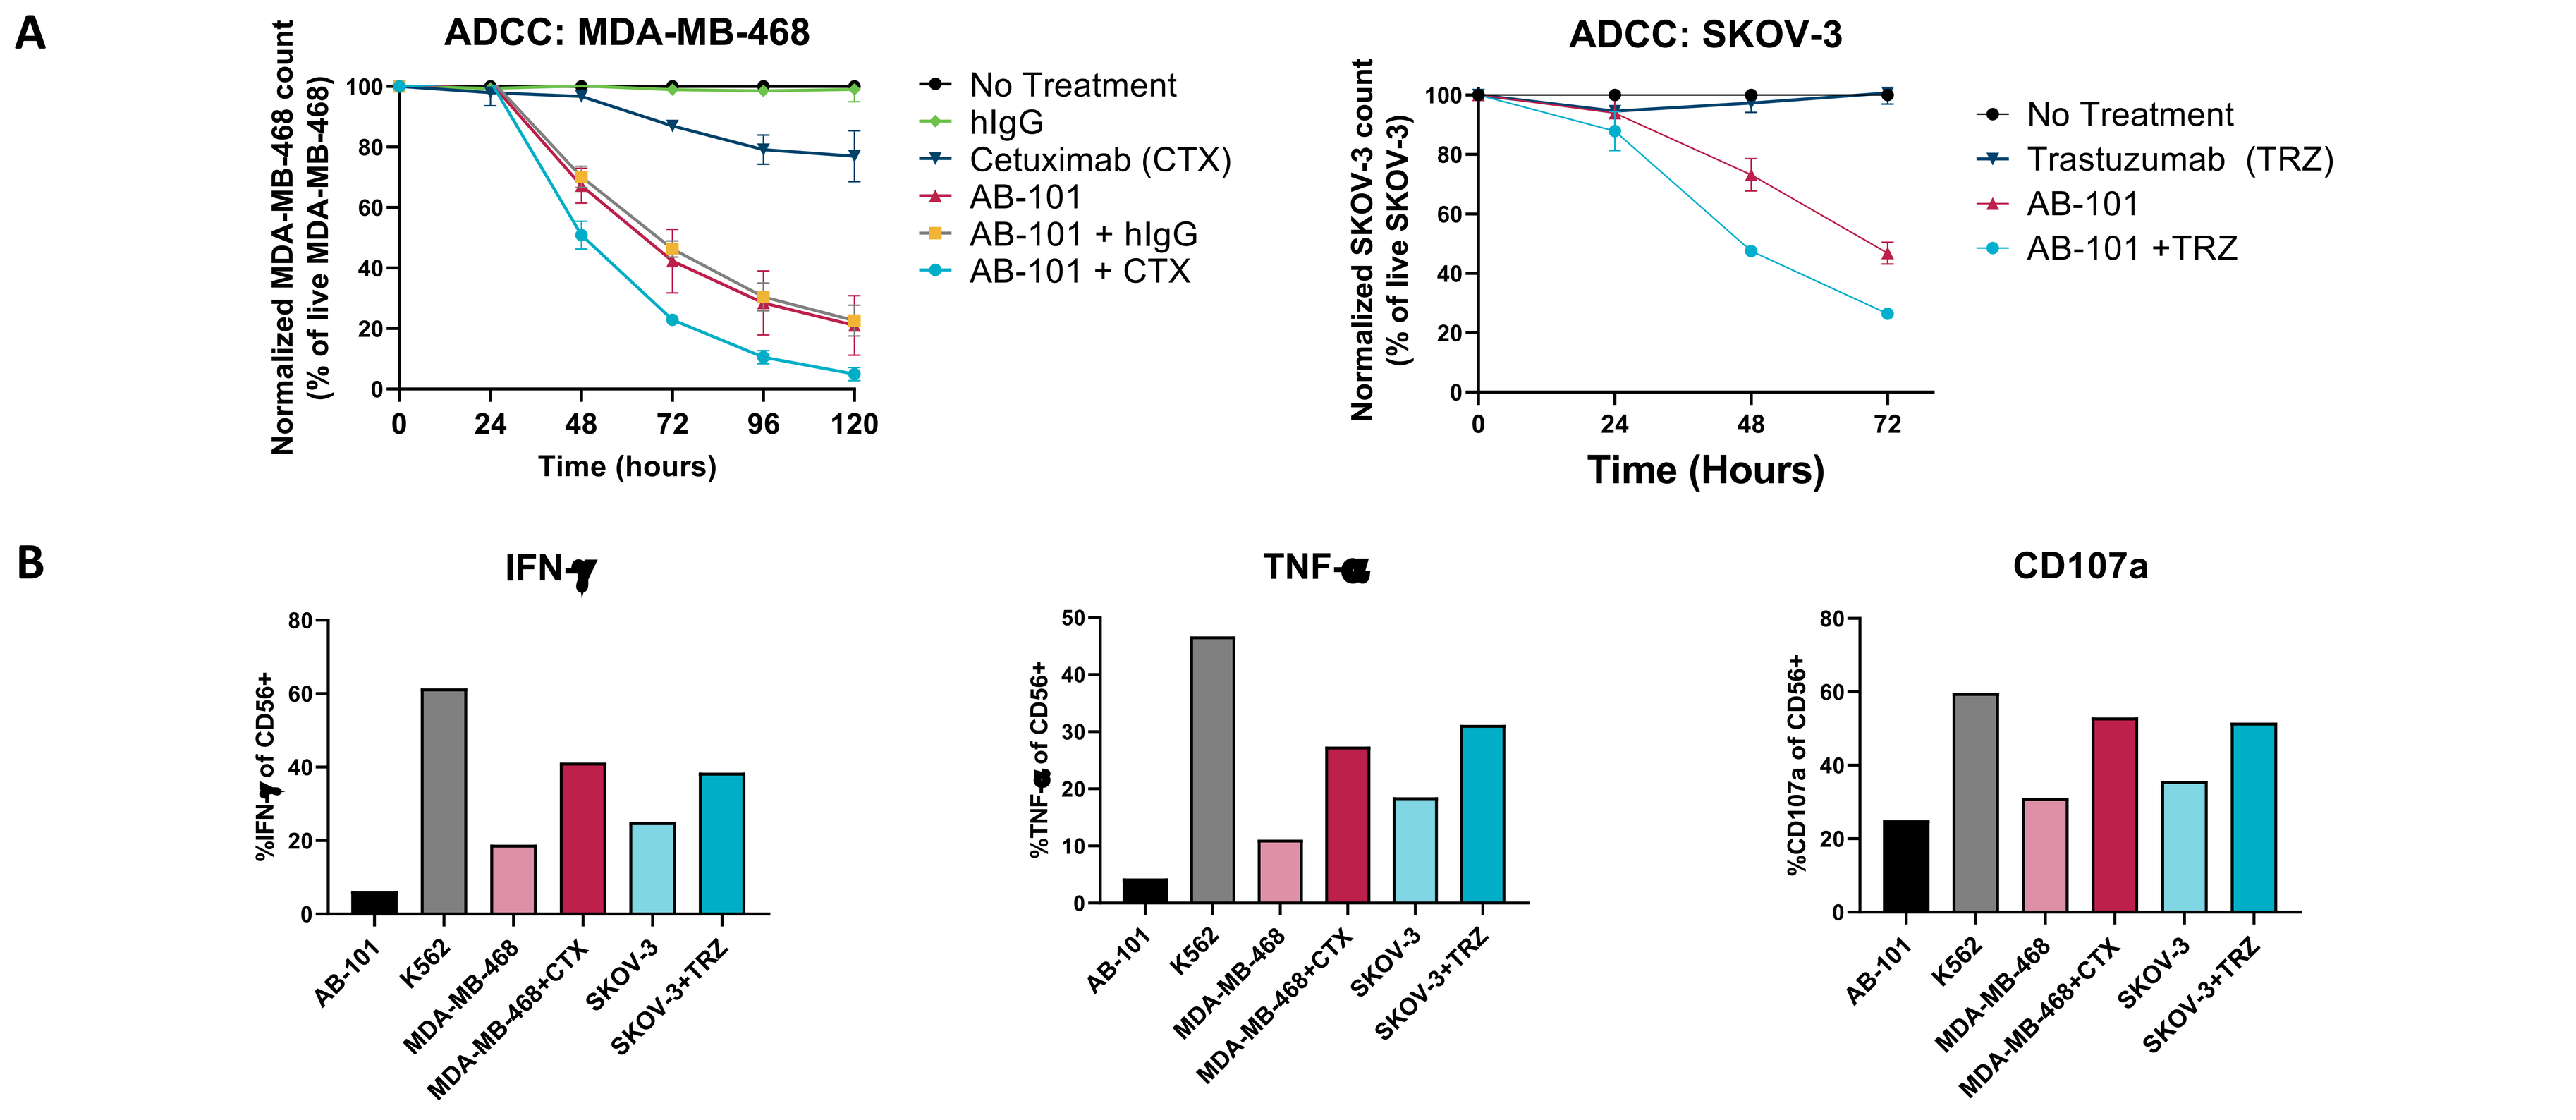
A
ADCC: MDA-MB-468
ADCC: SKOV-3
No Treatment
hIgG
Cetuximab (CTX)
AB-101
AB-101 + hIgG
AB-101 + CTX
Trastuzumab (TRZ)
AB-101 +TRZ
Normalized MDA-MB-468 count (% of live MDA-MB-468)
Normalized SKOV-3 count (% of live SKOV-3)
Time (hours)
Time (Hours)
B
CD107a
%CD107a of CD56+
AB-101
K562
MDA-MB-468
MDA-MB-468+CTX
SKOV-3
SKOV-3+TRZ

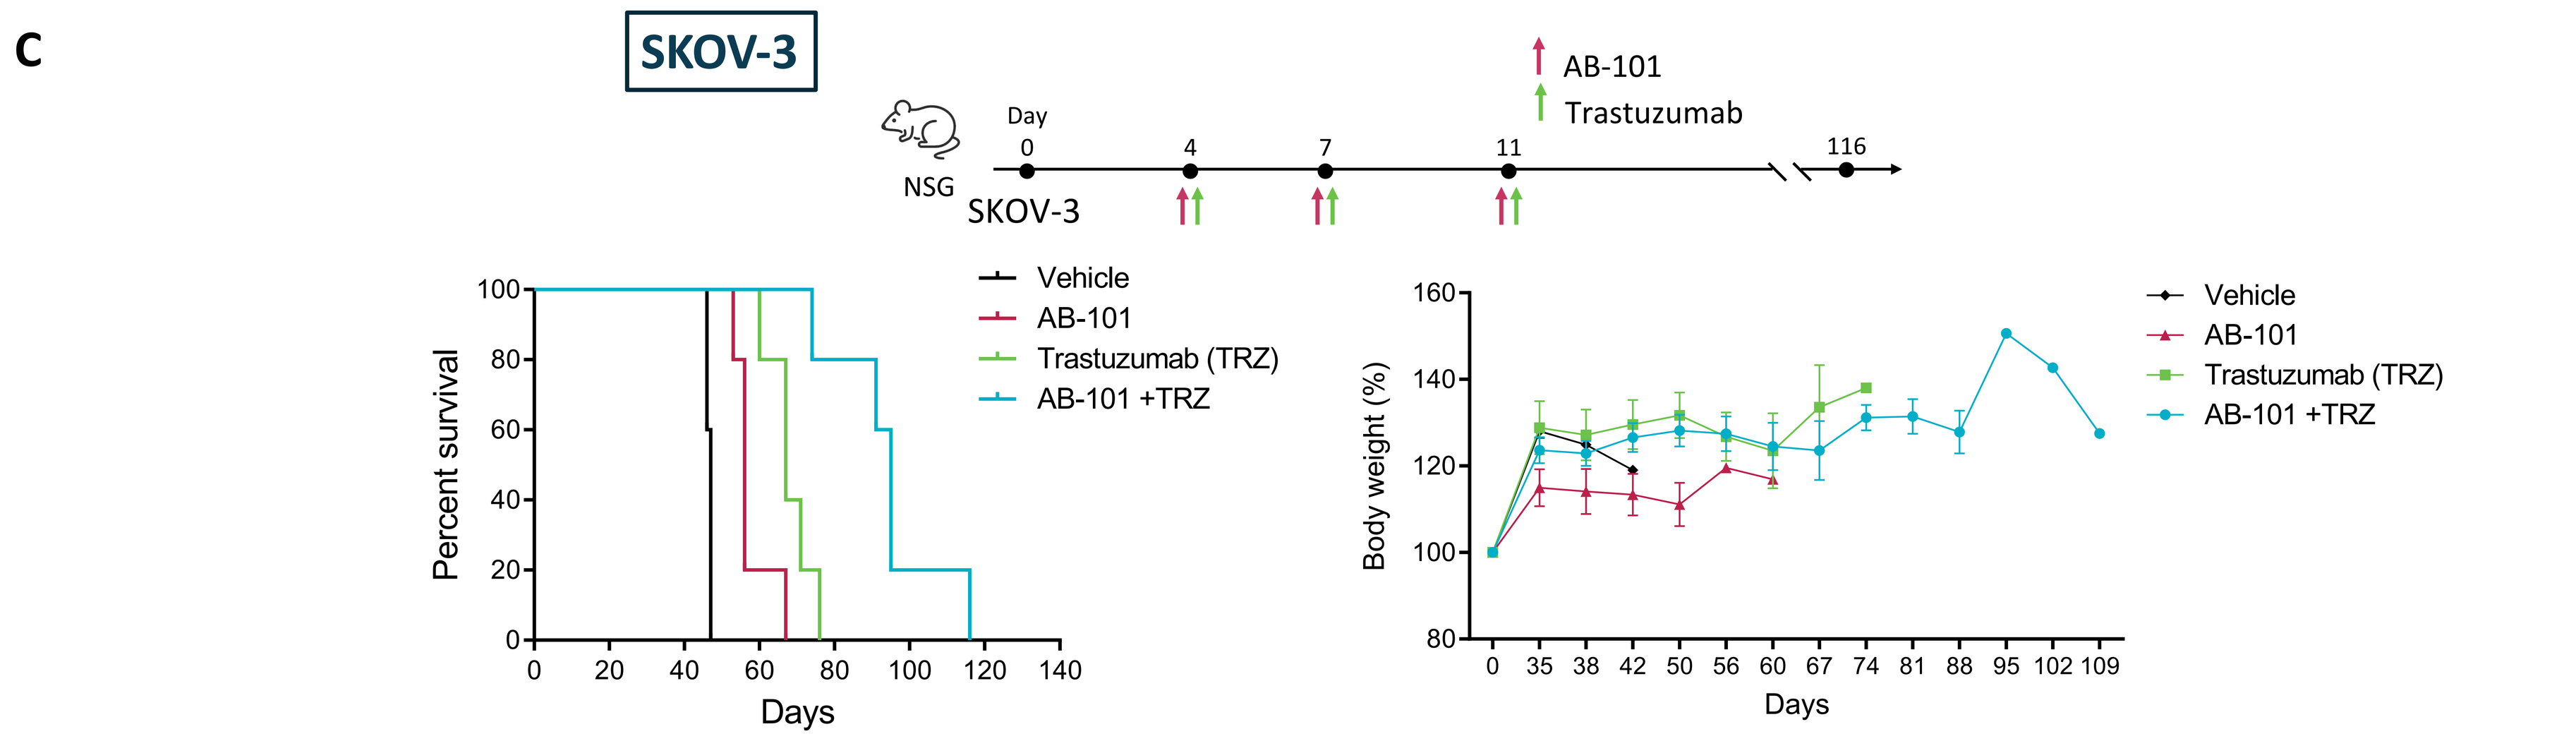
C
SKOV-3
AB-101
Trastuzumab
NSG
Day
116
Vehicle
AB-101
Trastuzumab (TRZ)
AB-101 +TRZ
Percent survival
Body weight (%)
Days

A. Long-term cellular cytotoxicity of AB-101 alone and in combination with Cetuximab (1µg/mL) or isotype control (hIgG) against an EGFR+ breast (MDA-MB-468) tumor cell line and AB-101 alone or in combination with Trastuzumab (1µg/mL) in HER2+ ovarian (SKOV-3) tumor cell line over 72-120 hours at a 3:1 E:T ratio (MDA-MB-468) or 1:1 E:T ratio (SKOV-3). Data are plotted as the mean ± SEM.
B. AB-101 was stimulated for 4 hours with tumor targets at a 1:1 E:T ratio. Following stimulation, cells were collected, and degranulation (CD107a) and cytokine secretion were analyzed by flow cytometry.
C. Experimental design: $1x10^6$ SKOV-3 tumor cells were inoculated by intraperitoneal (IP) administration on day 0 in NSG mice, followed by IP administration of vehicle, Trastuzumab (5mg/kg) , AB-101 ($2x10^7$), or AB-101 + Trastuzumab on day 4, 7, 11. Kaplan-Meier survival curve is representative of % survival in each group (left). No differences were observed in body weight between groups (right).

Conclusions

Data presented herein suggests that AB-101, a highly scaled, cryopreserved, allogeneic NK cell product, has potential to be an effective therapy in combination with mAbs as an ADCC enhancer in hematologic malignancies and solid tumor indications.

References

1. Cooley, S., et al., Donors with group B KIR haplotypes improve relapse-free survival after unrelated hematopoietic cell transplantation for acute myelogenous leukemia. Blood, 2009. 113(3): p. 726-32.
2. Musolino, A., et al., Immunoglobulin G fragment C receptor polymorphisms and clinical efficacy of trastuzumab-based therapy in patients with HER-2/neu-positive metastatic breast cancer. J Clin Oncol, 2008. 26(11): p. 1789-96.